Do countries with similar GDPs and health expenditures reimburse the same cancer drugs?

Jan McKendrick,[1] Bill Malcolm,[2] Kate Sheahan,[3] Ioannis Katsoulis,[1] Xirong Song,[1] Jeanni van Loon[1]

[1]PRMA Consulting Ltd, Fleet, UK; [2]Bristol-Myers Squibb Pharmaceuticals Ltd, Uxbridge, UK; [3]Gillings School of Global Public Health, University of North Carolina at Chapel Hill, Chapel Hill, NC, USA

Background and objectives

- In today's economically constrained environment, payers have responded to the potentially high cost of new oncology medicines by conducting value-for-money assessments through national or regional pricing and reimbursement (P&R) and health technology assessment (HTA) agencies.
- Assessments may introduce the potential for discrepancies between the clinically eligible population (under the license) and the population that is eligible for treatment through public reimbursement.
- This study analyzed whether gross domestic product (GDP) per capita (GDPPC) and health expenditure as a proportion of GDP are associated with the national access restrictions imposed in various markets.

Methods

- Six common cancers were identified (breast, kidney, lung, melanoma, multiple myeloma, and prostate). New oncology therapies granted a first license by the European Medicines Agency (EMA), Health Canada (HC) or the Australian Therapeutic Goods Administration (TGA) between January 2006 and June 2016 were identified along with any relevant follow-on indications.
- For each first regulatory approval, HTA/reimbursement decisions by national agencies were identified for the initial and follow-on indications.
- Decisions were classified according to the level of access restriction in relation to the clinically eligible population (Figure 1):

No restriction (unrestricted access to all patients)
Partial restriction (restricted to subpopulations)
Complete restriction (restricted – no patient access)

Figure 1: Patient populations

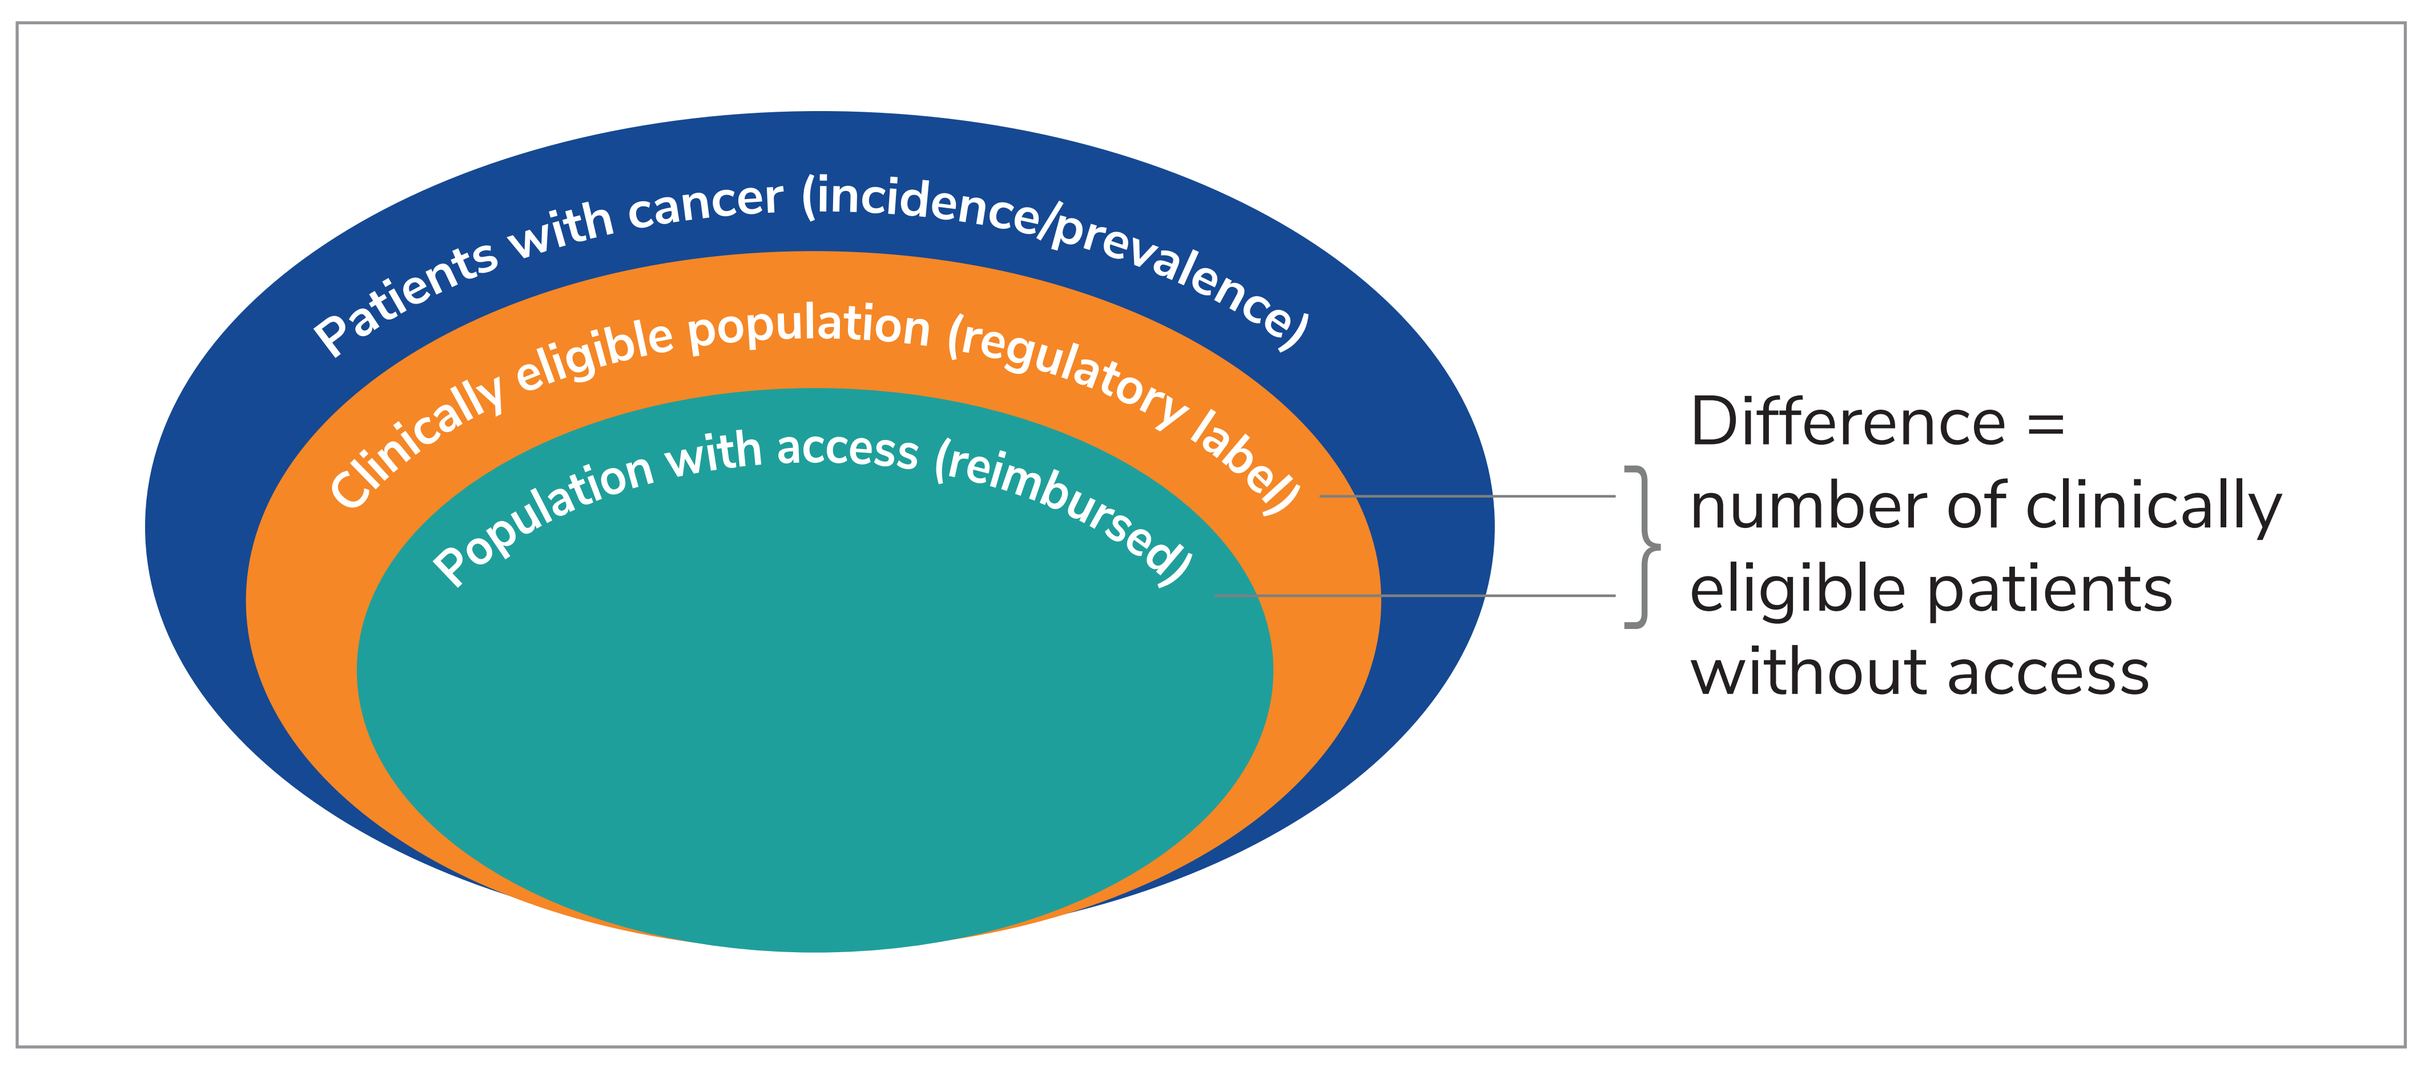
Patients with cancer (incidence/prevalence)
Clinically eligible population (regulatory label)
Population with access (reimbursed)
Difference = number of clinically eligible patients without access

Indicative graphic representation; the population sizes may vary substantially depending on the cancer type and intervention.

- Analyses are based on publicly available national HTA/P&R documents. Where no relevant documents were identified, other publicly available information and communication with country-level experts were used to ratify the reimbursement status. Where no conclusions could be drawn, the outcome of the assessment was considered to result in no restriction.
- Using the estimated median OS gain with the new treatment (from the EMA regulatory documents), potential years of life lost (YLL) as a result of any restrictions were estimated.
- For each reimbursement outcome, the impact of the decision was established in terms of the number of patients without access and YLL (assuming each assessment to be independent).
- The relationship between restrictions (nature and impact on patients) and two financial metrics for each country were explored (Table 1):
 - GDPPC – mean over 2006–2016[1]
 - health expenditure as a % of GDP – mean over 2006–2015.[2]

Table 1: Methods for assessing relationship between reimbursement restrictions and financial metrics

Outcome variables (per country)	Explanatory variable	Method
• Percentage of reimbursement outcomes that resulted in restriction (complete or partial) • Total number of patients impacted (per 100,000 population) • Total YLL (per 100,000 population)	• GDPPC • Health expenditure as a % of GDP	• Descriptive • Spearman rank correlation coefficient • Regression analysis (linear)

Results

Reimbursement decisions

- Of the 892 possible reimbursement decisions (Figure 2), 316 (35%) resulted in a complete or partial restriction (Table 2).

Figure 2: Reimbursement decisions included

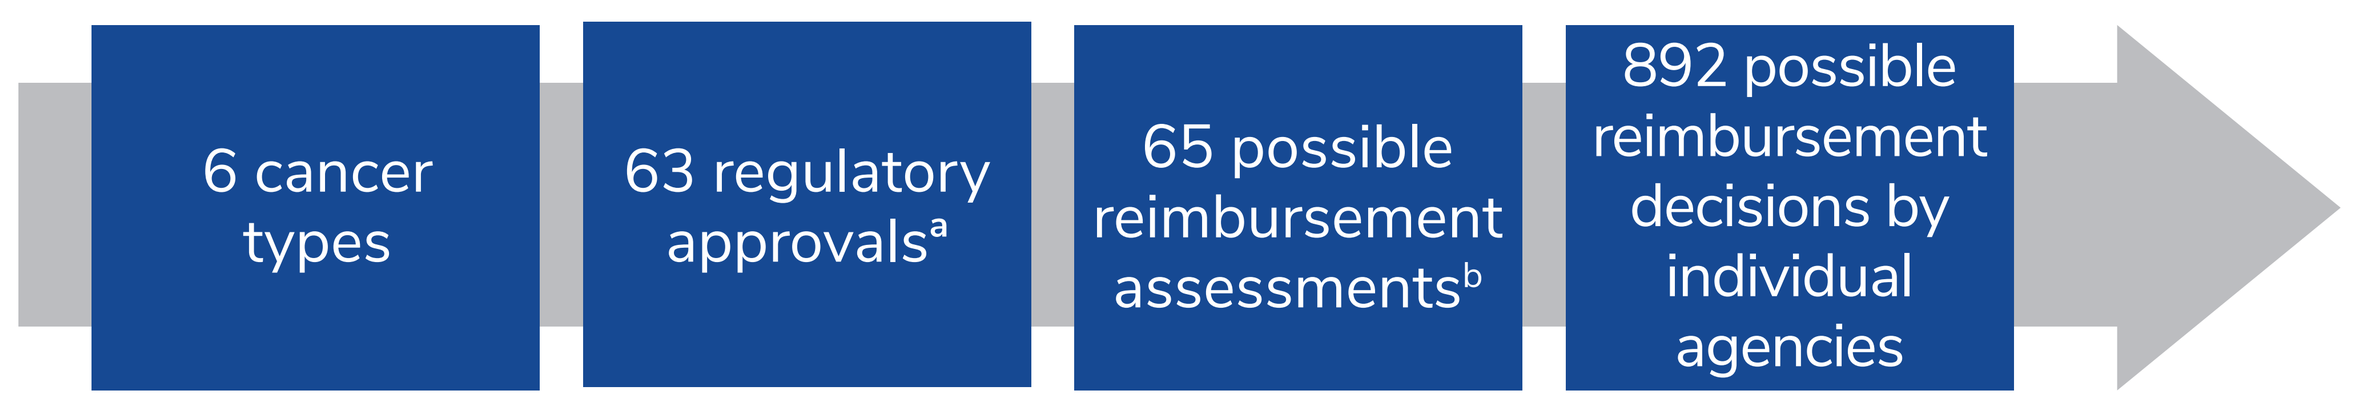
6 cancer types
63 regulatory approvals[a]
65 possible reimbursement assessments[b]
892 possible reimbursement decisions by individual agencies

[a] 56 regulatory approvals in Canada and Australia
[b] 2 regulatory decisions, which each led to 2 reimbursement decisions

Figure 2: Reimbursement decisions and their impact

Country	Complete or partial restrictions (%)	Impact per 100,000 population[a, b]	
		Patients without access through public reimbursement	YLL
Australia	64	104.4	32.7
Belgium	20	51.3	12.8
Canada	39	42.7	21.8
Denmark	37	107.1	18.1
France	17	33.5	5.9
Germany	0	0.0	0.0
Italy	37	81.8	20.0
Netherlands	3	9.4	2.4
Poland	98	136.1	39.4
Portugal	60	131.6	36.1
Spain	15	5.7	1.3
Sweden	14	75.6	34.6
UK	42[c], 54[d]	94.8	24.76

[a] GLOBOCAN 2012 data; [b] Assumptions: 100% market uptake for every drug/indication; each reimbursement assessment is independent; assessments can be added together to estimate impact; impact in Germany is assumed to be zero, as all approvals are in line with the license; [c] NICE; [d] SMC

- NICE and the SMC were similar in their decisions to restrict access to therapies and in the nature of the restrictions.
- Partial restrictions in Belgium and most in Canada, Denmark, Poland, and Spain were considered "refinements" of the licensed population based on inclusion/exclusion criteria from the product's pivotal trial(s).
- The most common refinement was restricting reimbursement based on an Eastern Cooperative Oncology Group (ECOG) performance status of 0–1 or 0–2. It was often unclear why a specific refinement had been imposed or what impact it would have in terms of the identification of patients in clinical practice.

Relationship between reimbursement restrictions and financial metrics

- As shown in Figure 3A, Poland and Portugal, with lower GDPPC, applied complete restrictions in 94% and 60% of assessments, respectively. However, Spain, with a somewhat higher but still relatively low GDPPC, applied a lower rate of complete restrictions (2%) at the national level; however, regional restrictions were not taken into account.
- Rates of complete restrictions varied among countries with similar GDPPC, such as Australia, Belgium, Canada, and Germany (0–31% of assessments).
- Analyses based on health care expenditure (Figure 3B) showed:
 - Germany, France, and the Netherlands, with ≥10% health expenditure as a % of GDP, imposed restrictions in 0–17% of indications.
 - Countries with <9% health expenditure as a % of GDP (e.g., Poland, Australia, Italy, and the UK) restricted access to a varying degree (37–98% of indications).

Figure 3: Degree of restriction and (A) GDPPC, (B) health expenditure as a % of GDP

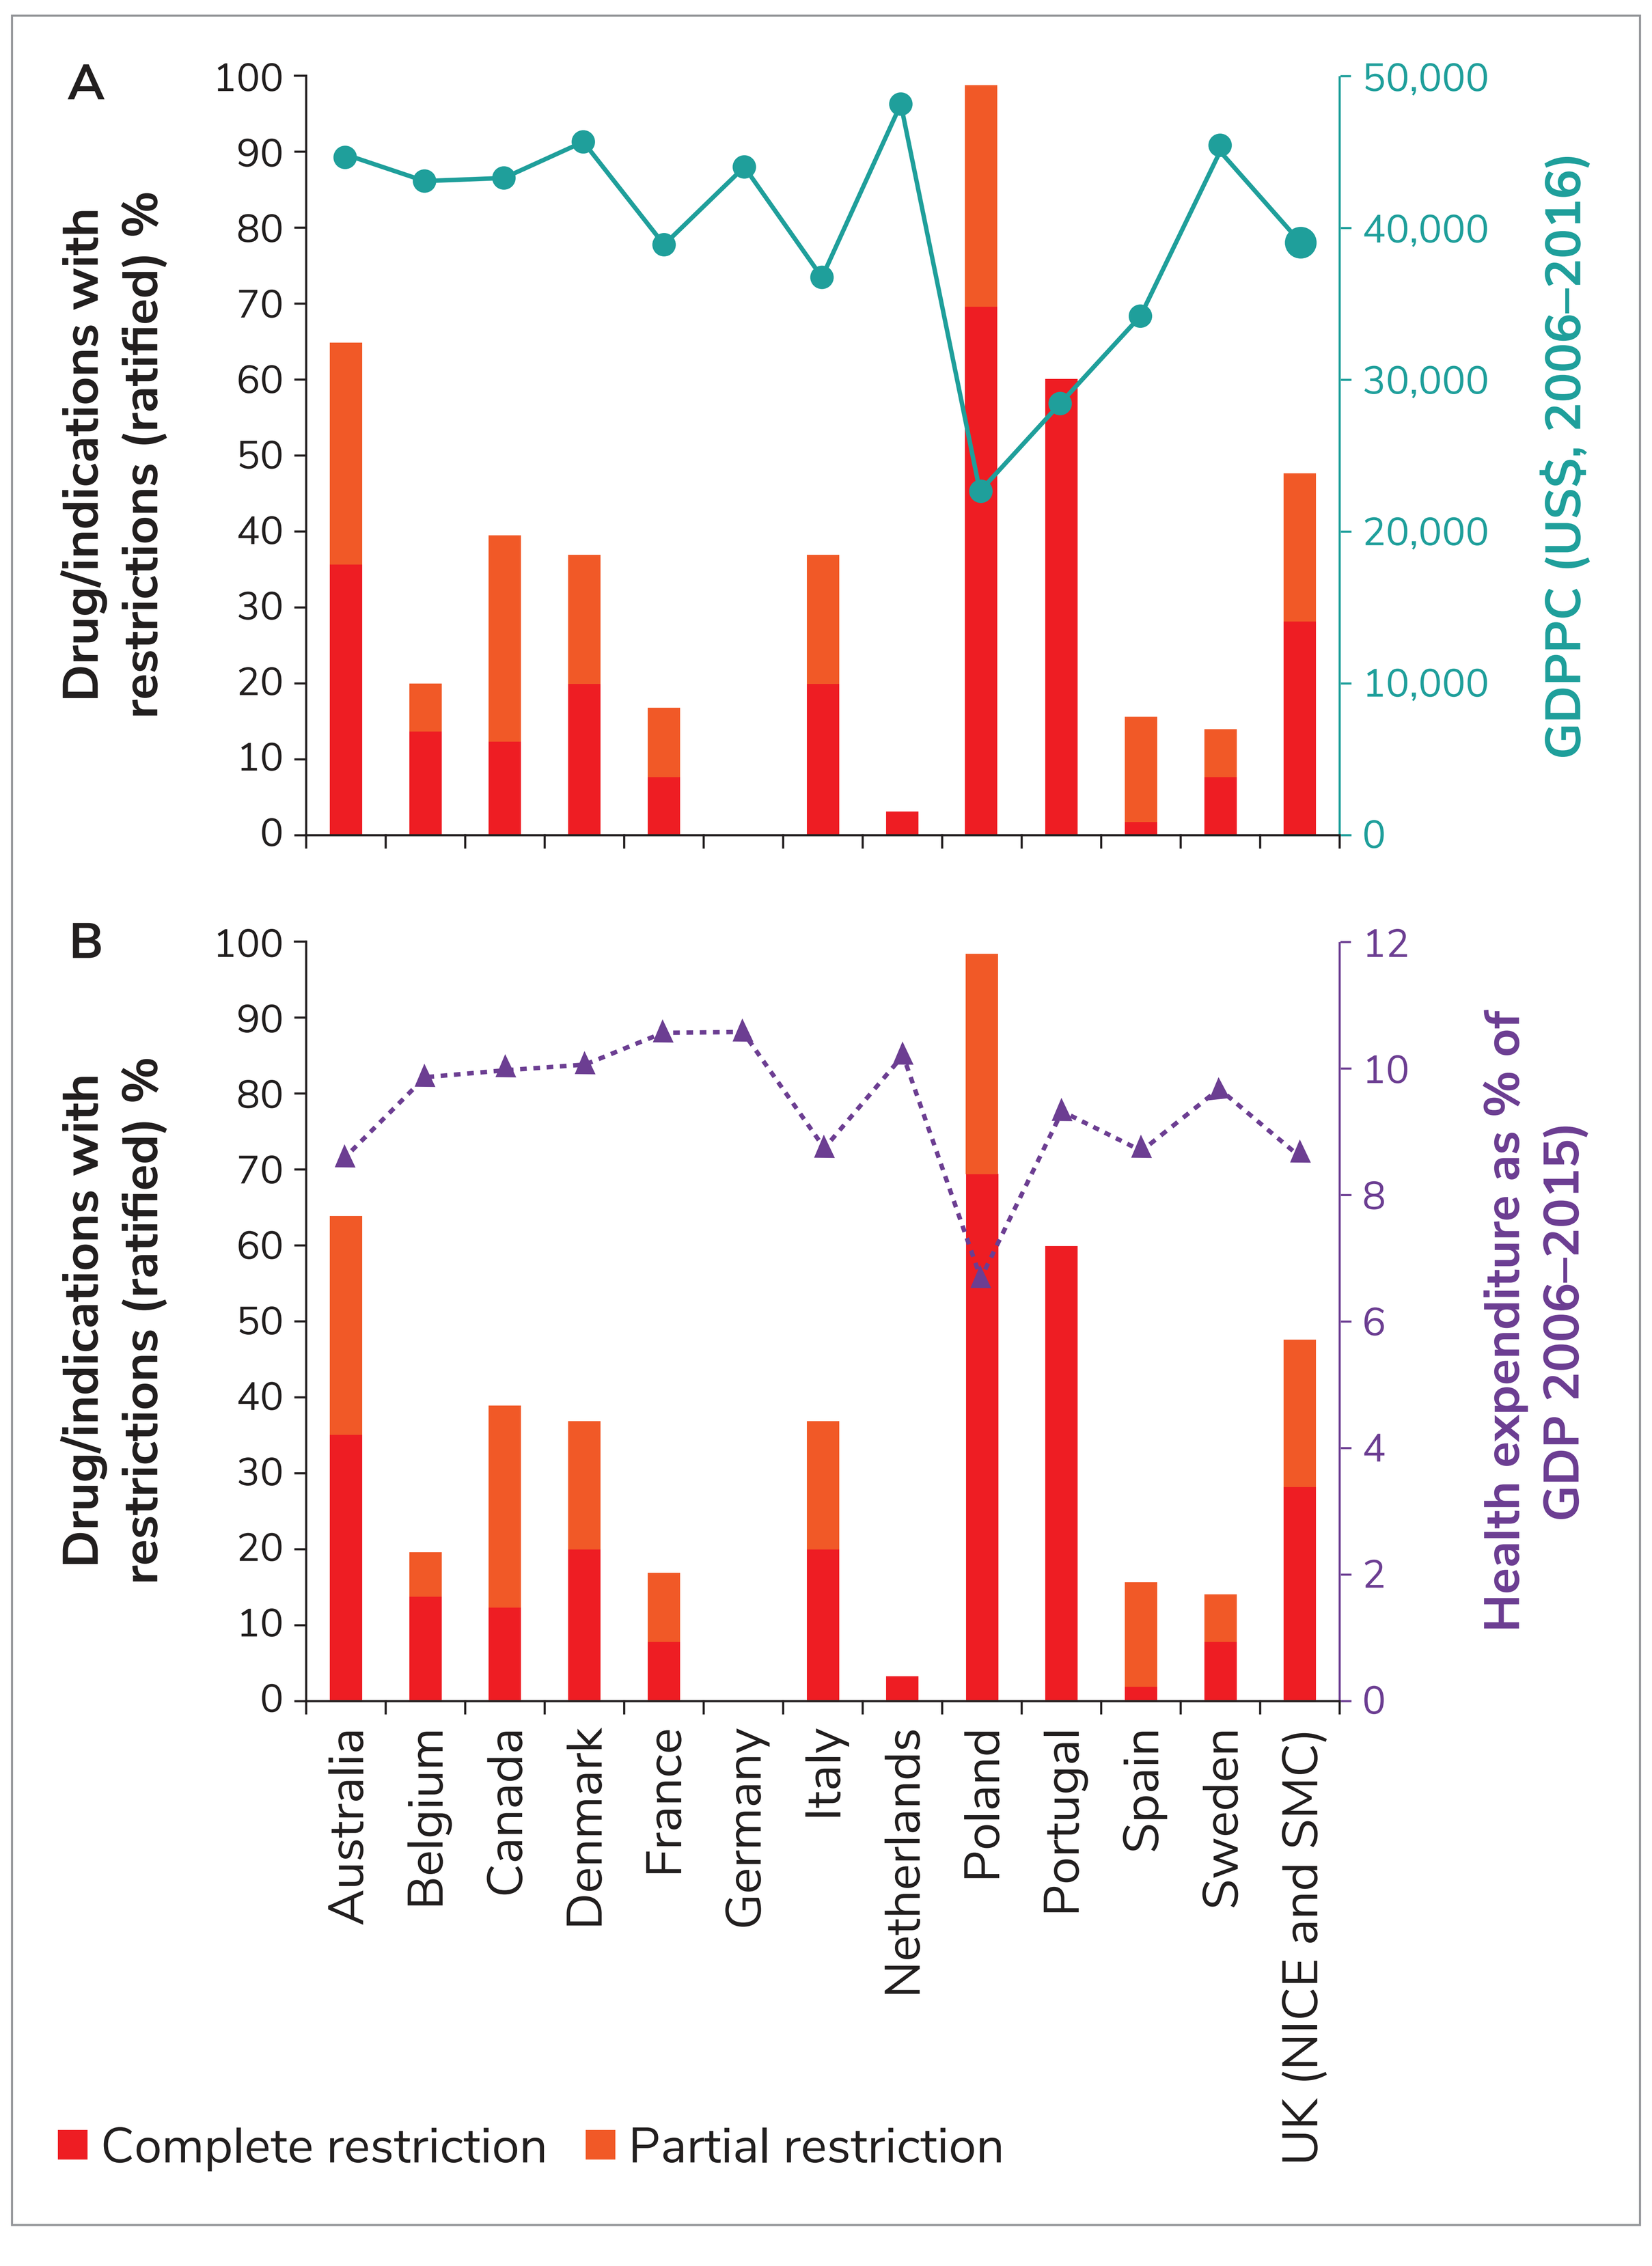
A
Drug/indications with restrictions (ratified) %
GDPPC (US$, 2006–2016)
B
Drug/indications with restrictions (ratified) %
Health expenditure as % of GDP 2006–2015)
Australia
Belgium
Canada
Denmark
France
Germany
Italy
Netherlands
Poland
Portugal
Spain
Sweden
UK (NICE and SMC)
Complete restriction
Partial restriction

- Although the rank correlation between the proportion of assessments with restrictions in each country and GDPPC was −0.71, the relationship was not linear (R^2 = 0.51, Table 3); however, there was a stronger linear relationship between health expenditure as a proportion of GDP and the proportion of reimbursement restrictions in each country (R^2 = 0.70, Table 3).

Table 3: Results of regression analyses

	GDPPC		Health expenditure as a proportion of GDP	
	R^2	Correlation coefficient	R^2	Correlation coefficient
Complete and partial restrictions (%)	0.4532	-0.7148	0.6671	-0.8357
Total number of patients without access	0.2371	-0.4869	0.3789	-0.6156
Total YLL	0.1747	-0.4180	0.3665	-0.6054

Relationship between impact of reimbursement outcomes and country financial metrics

- There were no clear patterns of association between GDPPC and the total number of patients affected or YLL (Figure 4A and Table 3).
- Analyses suggest that the number of patients without access increases with health expenditure but the relationship is not linear (Figure 4B and Table 3).

Figure 4: Total number of patients without access and total YLL vs (A) GDPPC and (B) health expenditure as a % of GDP

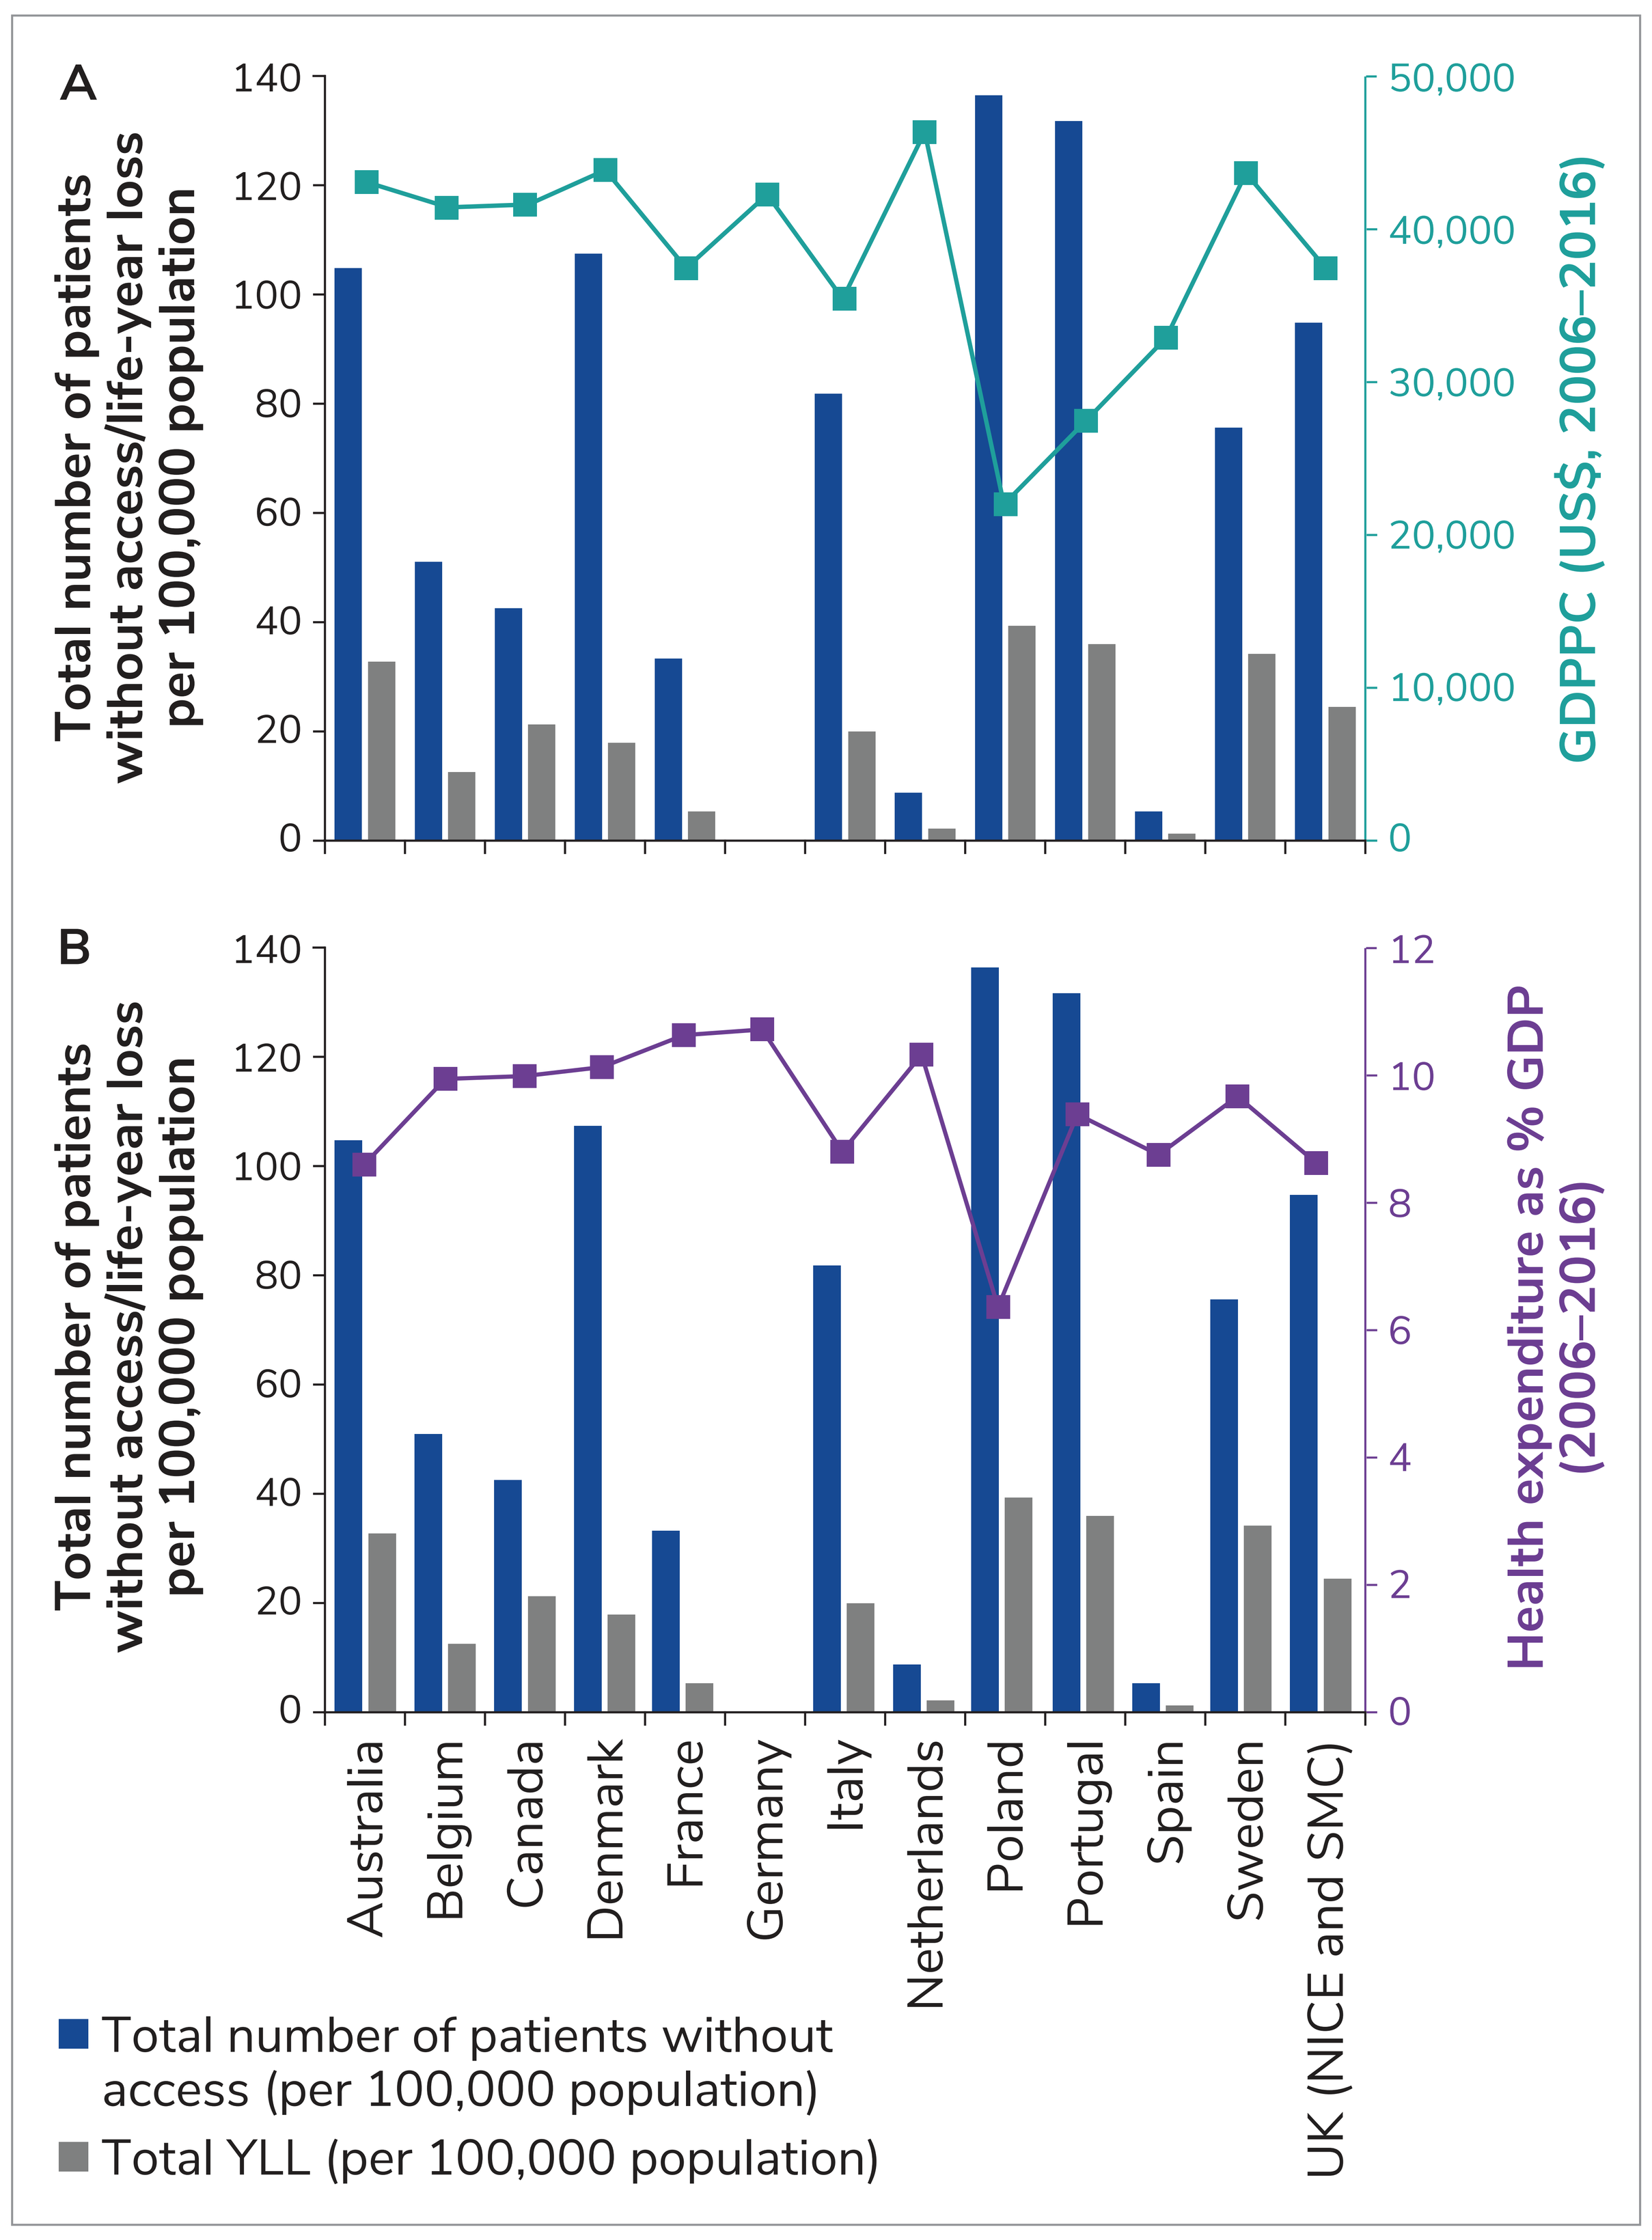
A
Total number of patients without access/life-year loss per 100,000 population
GDPPC (US$, 2006–2016)
B
Total number of patients without access/life-year loss per 100,000 population
Health expenditure as % GDP (2006–2016)
Australia
Belgium
Canada
Denmark
France
Germany
Italy
Netherlands
Poland
Portugal
Spain
Sweden
UK (NICE and SMC)
Total number of patients without access (per 100,000 population)
Total YLL (per 100,000 population)

Discussion and conclusions

- Access to cancer therapies via public reimbursement varies between countries, and the nature of the restrictions and their impact on patients is heterogeneous.
- Patterns of restrictions do not correlate with GDP; however, healthcare expenditure as a proportion of GDP was moderately predictive of the extent of restriction.
- Analysis of reimbursement outcomes (i.e., patients affected and YLL) with GDPPC and health expenditure as a proportion of GDP indicated that these correlated in terms of ranking, but the relationship could not easily be explained using linear regression models. **The findings suggest that, in the countries included in this study, inequitable access to cancer treatments resulting from discrepancies between the licensed and reimbursed populations cannot be explained by GDPPC.**

References

1. https://data.oecd.org/gdp/gross-domestic-product-gdp.htm
2. http://stats.oecd.org/Index.aspx?DataSetCode=SHA

Footnotes

This work was conducted by PRMA Consulting Ltd and funded by Bristol-Myers Squibb.